An Automated Lupus Anticoagulant Assay Resistant to Anticoagulant Drugs Including DOACs and Oral FXI Inhibitors

Abstract Number: PB1253

Mushfiqur Rahman, Karen M. Black, Ali Sadeghi-Khomami
Precision BioLogic Inc., Dartmouth, Nova Scotia, Canada

Presented at ISTH 2026
July 11–15, 2026

PrecisionBioLogic

Background

Direct oral anticoagulants (DOACs) and oral FXI inhibitors interfere with lupus anticoagulant (LA) testing by prolonging clotting times and increasing assay variability. Activated charcoal (Act-C) adsorbs hydrophobic anticoagulants but typically requires manual plasma sample pretreatment.[1-2]

Although the CRYO*check*™ Hex LA™ assay is resistant to heparin, vitamin K antagonists (VKAs), and factor deficiencies while remaining diagnostically accurate in the presence of DOACs and oral FXI inhibitors,[3-4] Act-C integration could further reduce anticoagulant-related clotting-time prolongation and delta value increase.

This study evaluated whether Act-C could be directly integrated into the CRYO*check* Hex LA assay to create an automated anticoagulant-resistant LA assay without the need for plasma sample pretreatment.

Objective

To evaluate the feasibility of an integrated lupus anticoagulant assay incorporating screening, confirmatory, and mixing test functionality that is resistant to anticoagulant drug interference without pretreatment of plasma sample.

Methods

Act-C (50 mg) was integrated into the LA-APTT reagent of the Hex LA assay and into the CRYO*check* LA Check™ and LA Sure™ dRVVT reagents.

Original and modified formulations were tested on a mechanical and an optical coagulation analyzer using lupus-negative and lupus-positive plasma samples, contrived with UFH, LMWH (2 IU/mL), VKA (INR 3.5), DOACs (apixaban, edoxaban, rivaroxaban, dabigatran; 500 ng/mL), FXI inhibitors (asundexian, milvexian) or fondaparinux (1000 ng/mL).

Results

Act-C was incompatible with optical coagulation analyzer (ACL TOP-700, Sysmex CS-2500) and the dRVVT assay but was successfully integrated into the Hex LA formulation on a Stago mechanical coagulation analyzer.

Act-C did not impair heparin neutralization, and diagnostic outcomes matched the original Hex LA while reducing clotting-time prolongation and non-diagnostic delta increases caused by anticoagulant drugs **(Table 1)**.

Resistance was observed across DOACs, oral FXI inhibitors, UFH, LMWH, fondaparinux, and VKA-contrived plasma without requiring plasma pretreatment.

Conclusions

Direct integration of activated charcoal into Hex LA, a two-step incubation lupus anticoagulant assay, allowed us to formulate the first LA assay resistant to interference from all major anticoagulants without a need to pretreat plasma samples.

Diagnostic performance and resistance to heparins, fondaparinux, and VKAs of Hex LA were preserved with Act-C integration on mechanical coagulation analyzers.

Act-C integration was incompatible with optical coagulometers and the dRVVT assay.

References

1. Emmanuel J. Favaloro, Leonardo Pasalic. Res Pract Thromb Haemost. 2022;6:e12676.
2. T. Exner, N. Michalopoulos, J. Pearce, R. Xavier, M. Ahuja. *Thromb Res*. 2018;163:117–122.
3. CRYO*check* Hex LA™ Instructions for Use. Precision BioLogic Inc.
4. Rahman M, et al. Presented at ISTH 2023.

Table 1

Comparison of Average Clotting Times and Delta Values Between the Original and Act-C Incorporated Hex LA Formulations Across Various Contrived Plasma Samples (N=2, STA-R Max 3, LA positivity defined as Δ > 6 sec).

		Original Hex LA			Act-C Hex LA		
	Sample	**Start**	**Correct**	**Delta**	**Start**	**Correct**	**Delta**
Untreated	LA -	49.72	51.35	-1.63	49.82	51.30	-1.48
	LA +	70.30	58.50	**11.80**	70.08	56.33	**13.75**
	LA ++	114.10	81.96	**32.14**	111.41	79.81	**31.60**
VKA Plasma (3.5 INR)	(VKA) LA -	55.80	57.92	-2.12	54.70	58.27	-3.56
	(VKA) LA +	ND	ND	ND	ND	ND	ND
	(VKA) LA ++	104.76	84.61	**20.15**	105.43	79.94	**25.49**
Unfractionated Heparin (2 IU/mL)	LA -	48.30	50.02	-1.72	48.08	48.42	-0.34
	LA +	65.91	55.85	**10.06**	66.26	55.98	**10.28**
	LA ++	103.85	75.75	**28.10**	107.83	78.63	**29.20**
Low Molecular Weight Heparin (2 IU/mL)	LA -	53.18	54.04	-0.86	51.81	56.07	-4.25
	LA +	74.90	63.44	**11.45**	76.21	64.18	**12.02**
	LA ++	121.59	86.70	**34.89**	121.37	90.02	**31.36**
Fondaparinux (1000 ng/mL)	LA -	51.40	51.96	-0.55	51.39	51.93	-0.54
	LA +	73.24	61.20	**12.04**	76.12	59.73	**16.40**
	LA ++	117.30	86.31	**31.00**	118.96	86.8	**32.16**
Apixaban (500 ng/mL)	LA -	56.95	58.00	-1.05	50.33	51.24	-0.91
	LA +	87.85	67.42	**20.43**	75.64	60.22	**15.41**
	LA ++	158.63	97.54	**61.10**	117.82	85.88	**31.94**
Edoxaban (500 ng/mL)	LA -	62.73	65.87	-3.14	49.82	51.76	-1.94
	LA +	107.60	81.82	**25.78**	74.11	58.94	**15.18**
	LA ++	191.21	121.65	**69.56**	114.40	80.17	**34.22**
Rivaroxaban (500 ng/mL)	LA -	69.63	71.19	-1.57	50.46	52.09	-1.64
	LA +	133.76	88.53	**45.24**	74.95	59.61	**15.34**
	LA ++	221.14	134.90	**86.24**	116.68	78.09	**38.59**
Dabigatran (500 ng/mL)	LA -	95.12	98.80	-3.67	55.14	56.03	-0.89
	LA +	196.99	130.81	**66.18**	82.99	65.04	**17.95**
	LA ++	323.49	200.05	**123.44**	131.45	96.42	**35.02**
Milvexian (1000 ng/mL)	LA -	67.99	70.94	-2.95	51.30	50.46	0.84
	LA +	107.91	83.88	**24.04**	76.56	63.85	**12.71**
	LA ++	166.30	115.92	**50.38**	117.29	87.22	**30.07**
Asundexian (1000 ng/mL)	LA -	56.04	59.23	-3.19	49.86	51.54	-1.68
	LA +	83.12	68.75	**14.38**	73.08	61.67	**11.40**
	LA ++	131.07	92.62	**38.45**	111.51	82.03	**29.48**

LA -: CRYO*check* Lupus Negative Control; **LA +:** CRYO*check* Weak Lupus Positive Control; **LA ++:** CRYO*check* Lupus Positive Control;
(VKA) LA -: Pool of 3 patients' plasma (3.5 INR); **(VKA) LA ++:** 15% (v/v) of a strong LA antibody serum spiked in (VKA) LA -; **ND:** Not Determined.

Figure 1

LA-Negative Plasma Spiked with Anticoagulants: Control vs Act-C Hex LA Formulations

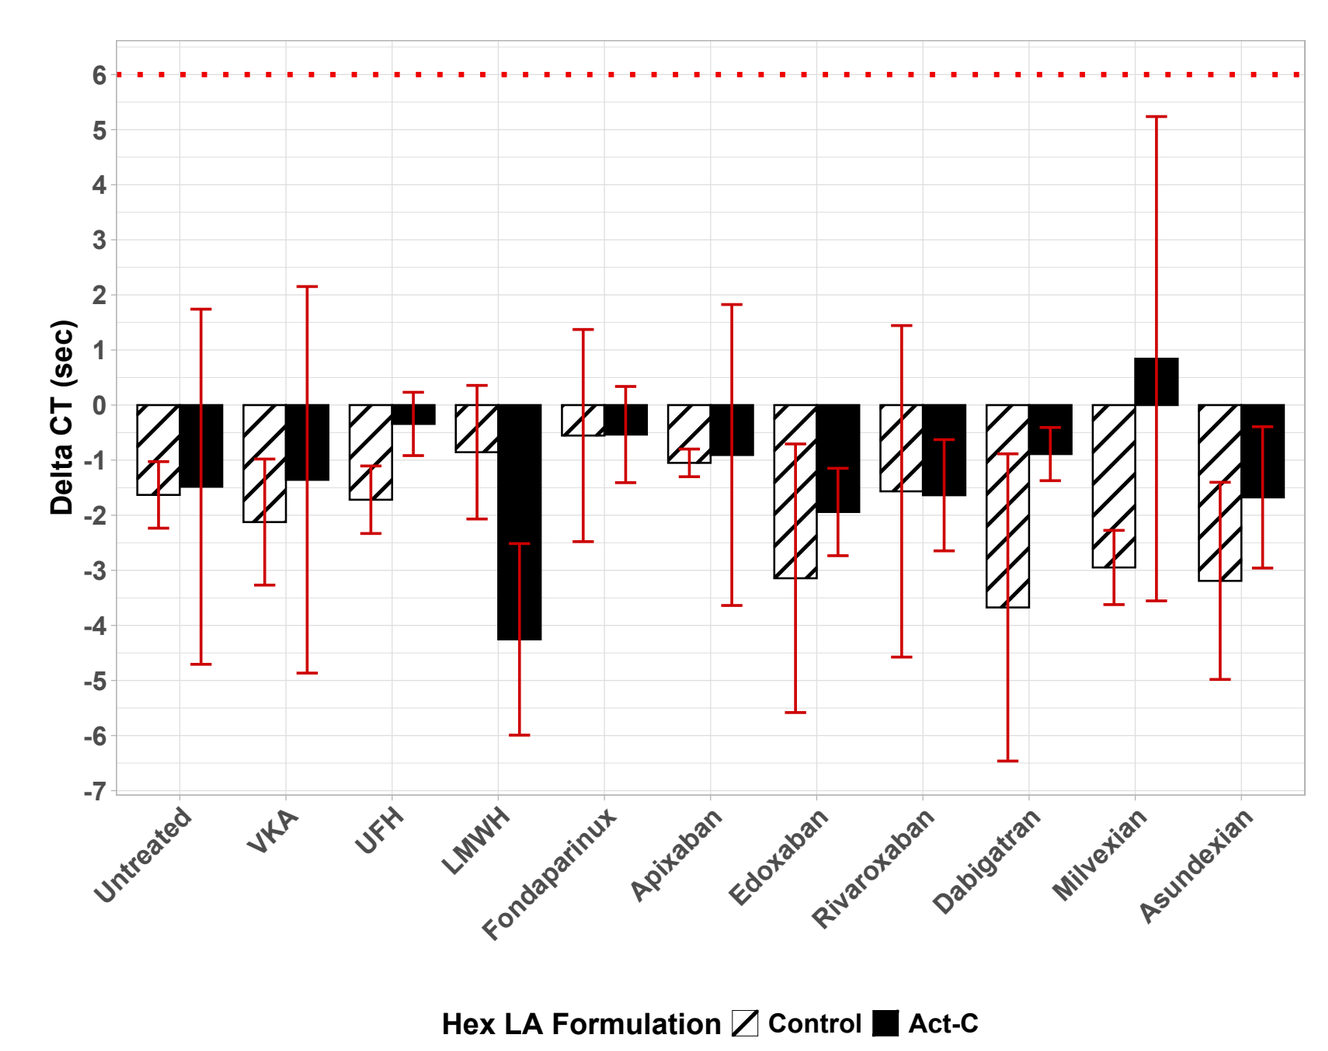
Delta CT (sec)
Untreated
VKA
UFH
LMWH
Fondaparinux
Apixaban
Edoxaban
Rivaroxaban
Dabigatran
Milvexian
Asundexian
Hex LA Formulation Control Act-C

(Figures 1–3) Comparison of Delta Clotting Times Between Original and Act-C Incorporated Hex LA Formulations in LA-Negative, LA-Positive, and Strong LA-Positive Plasma Spiked with Anticoagulants (STA-R Max 3, N = 2; ΔCT cutoff = 6 sec).

Patterned bars represent the original Hex LA formulation, and solid black bars represent the Act-C incorporated formulation.

Anticoagulant conditions included direct oral anticoagulants (apixaban, edoxaban, rivaroxaban, and dabigatran; 500 ng/mL), FXI inhibitors (asundexian and milvexian; 1000 ng/mL), unfractionated heparin (UFH; 2 IU/mL), low-molecular-weight heparin (LMWH; 2 IU/mL), fondaparinux (1000 ng/mL), and VKA-contrived plasma (INR 3.5).

Error bars represent SD. The red dashed line indicates the ΔCT positivity cutoff (6 sec).

Figure 2

LA-Positive Plasma Spiked with Anticoagulants: Control vs Act-C Hex LA Formulations

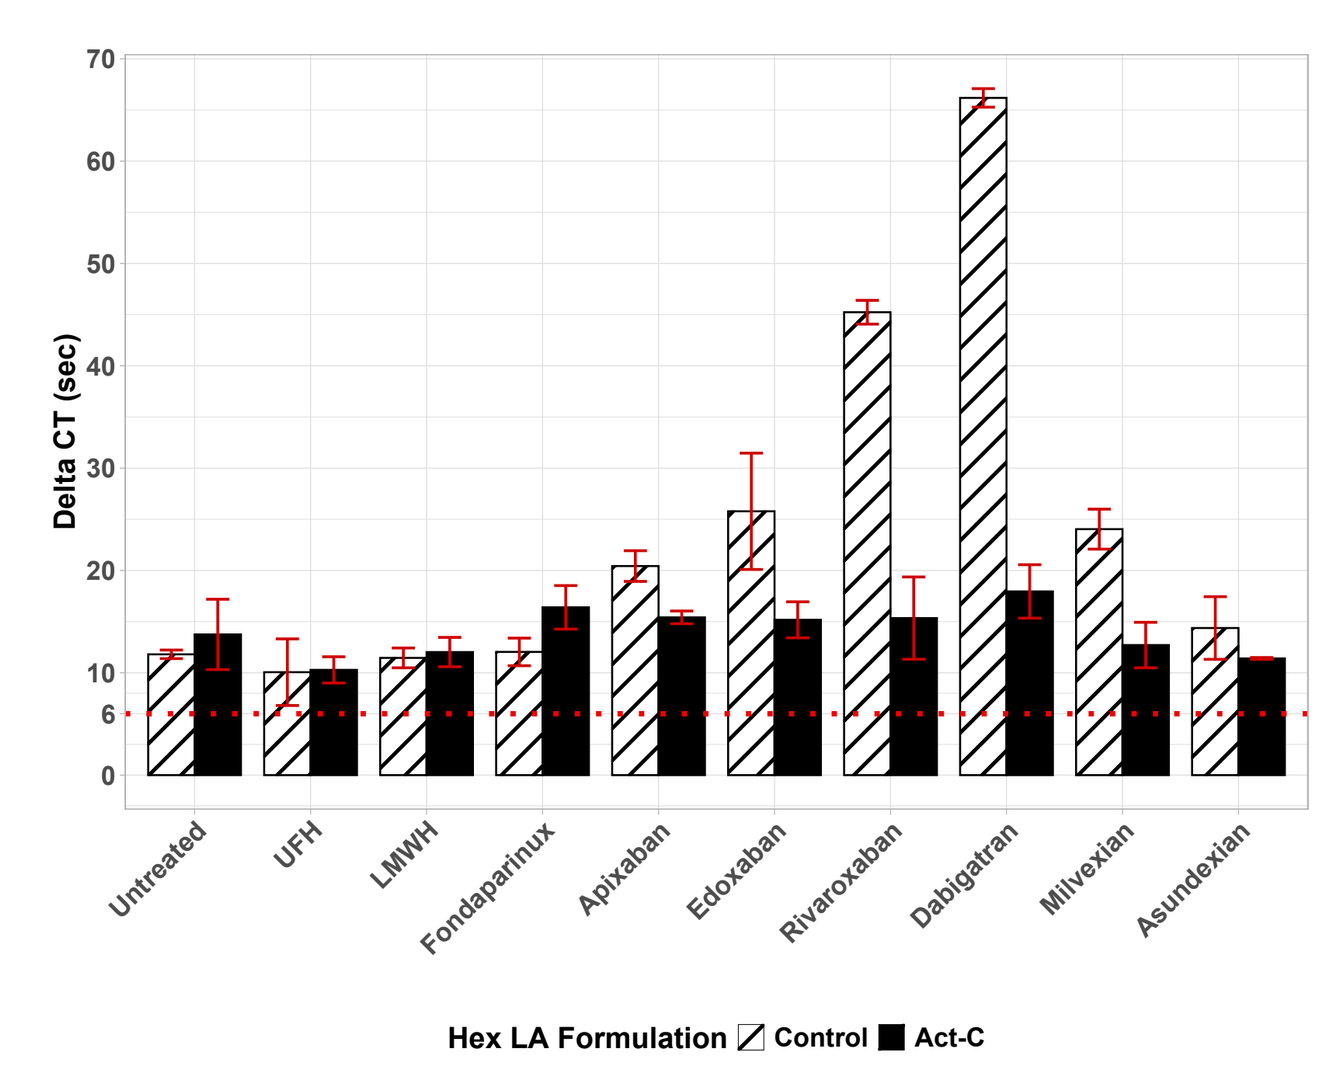
Delta CT (sec)
Untreated
UFH
LMWH
Fondaparinux
Apixaban
Edoxaban
Rivaroxaban
Dabigatran
Milvexian
Asundexian
Hex LA Formulation Control Act-C

Figure 3

Strong LA-Positive Plasma Spiked with Anticoagulants: Control vs Act-C Hex LA Formulations

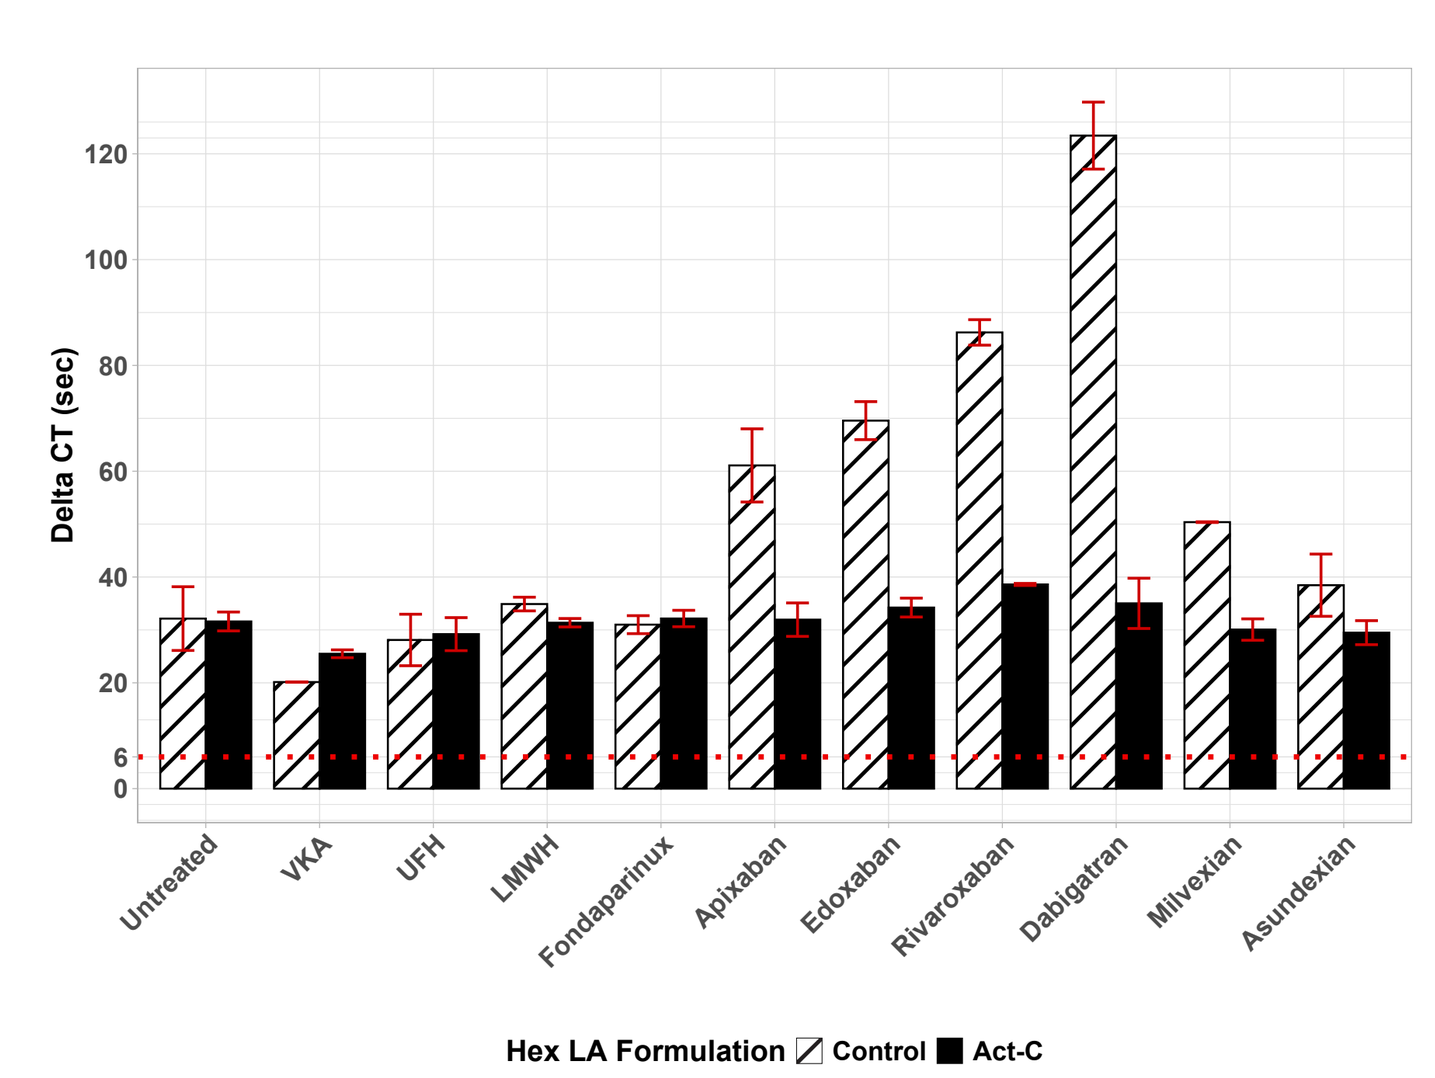
Delta CT (sec)
Untreated
VKA
UFH
LMWH
Fondaparinux
Apixaban
Edoxaban
Rivaroxaban
Dabigatran
Milvexian
Asundexian
Hex LA Formulation Control Act-C

Figure 4

Act-C Integration Reduces Anticoagulant-associated Analytical Clotting Time Prolongation in the Hex LA Formulation

Mean clotting times obtained with the original Hex LA formulation (left panels) and the Act-C incorporated Hex LA formulation (right panels) across LA-negative, LA-Positive, and Strong LA-Positive plasma samples contrived with various anticoagulants.

LA Start clotting times are shown as teal circles and trend lines, while LA Correct clotting times are shown as brown squares and trend lines.

Anticoagulant conditions included UFH, LMWH, fondaparinux, VKA, asundexian, apixaban, edoxaban, milvexian, rivaroxaban, and dabigatran.

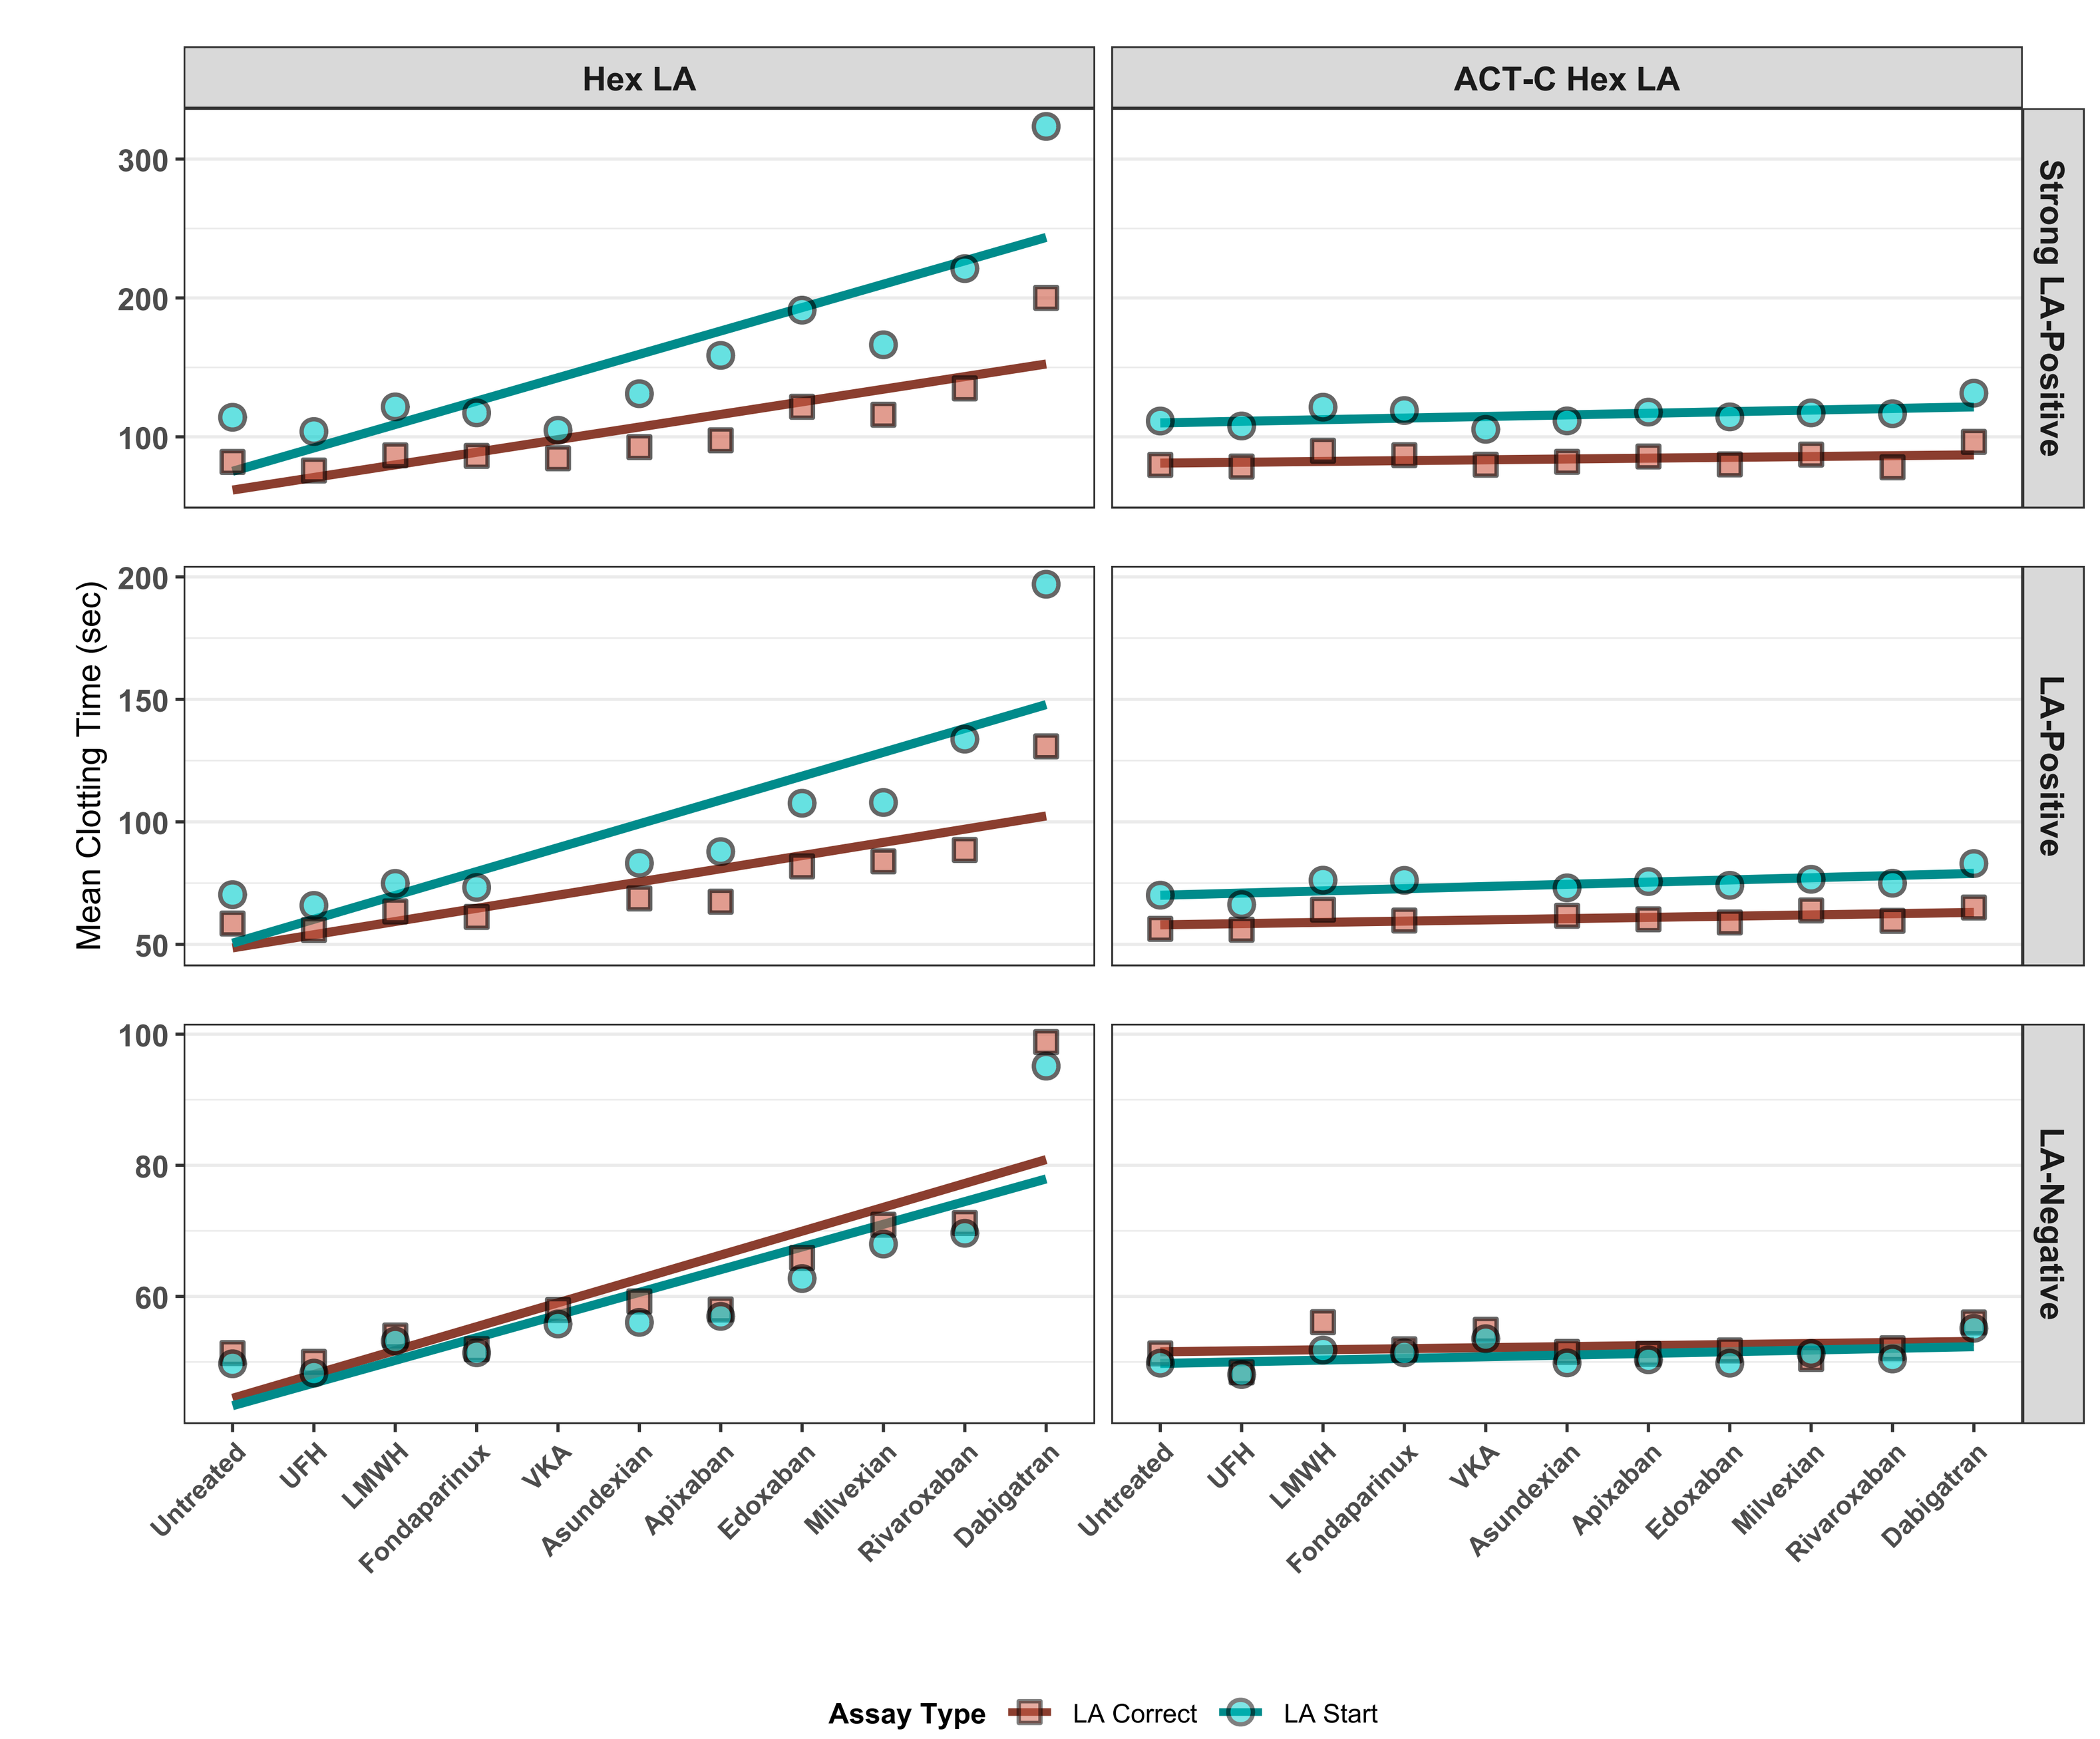
Hex LA
ACT-C Hex LA
Strong LA-Positive
LA-Positive
LA-Negative
Mean Clotting Time (sec)
Untreated
UFH
LMWH
Fondaparinux
VKA
Asundexian
Apixaban
Edoxaban
Milvexian
Rivaroxaban
Dabigatran
Assay Type LA Correct LA Start